#1033P: The phase I INSIGHT platform trial: Strata A and B evaluating feasibility of intratumoral and intraperitoneal IMP321 (soluble LAG-3 protein, eftilagimod alpha) in advanced solid tumors

Salah-Eddin Al-Batran[1,3], Daniel W. Mueller[1,3], Mohammad-Reza Rafiyan[2], Dragan Kiselicki[2], Timursah Habibzade[2], Chrystelle Brignone[4], Marina Schaaf[3], Regina Eickhoff[3], Elke Jäger[2], Thorsten O. Goetze[1,3]

[1]Krankenhaus Nordwest, University Cancer Center Frankfurt, Frankfurt, Germany; [2]Krankenhaus Nordwest, Frankfurt, Germany; [3]Institut für Klinische Krebsforschung IKF GmbH am Krankenhaus Nordwest, Frankfurt, Germany; [4]Immutep SAS, Orsay, France

Background

Strata A/B of the INSIGHT study evaluate feasibility of intratumoral (i.t.) and intraperitoneal (i.p.) eftilagimod alpha (efti, IMP321) monotherapy in advanced solid tumors. This MHC class II agonist activates antigen presenting cells followed by CD8 T- cell activation.

Figure 1: Study Design

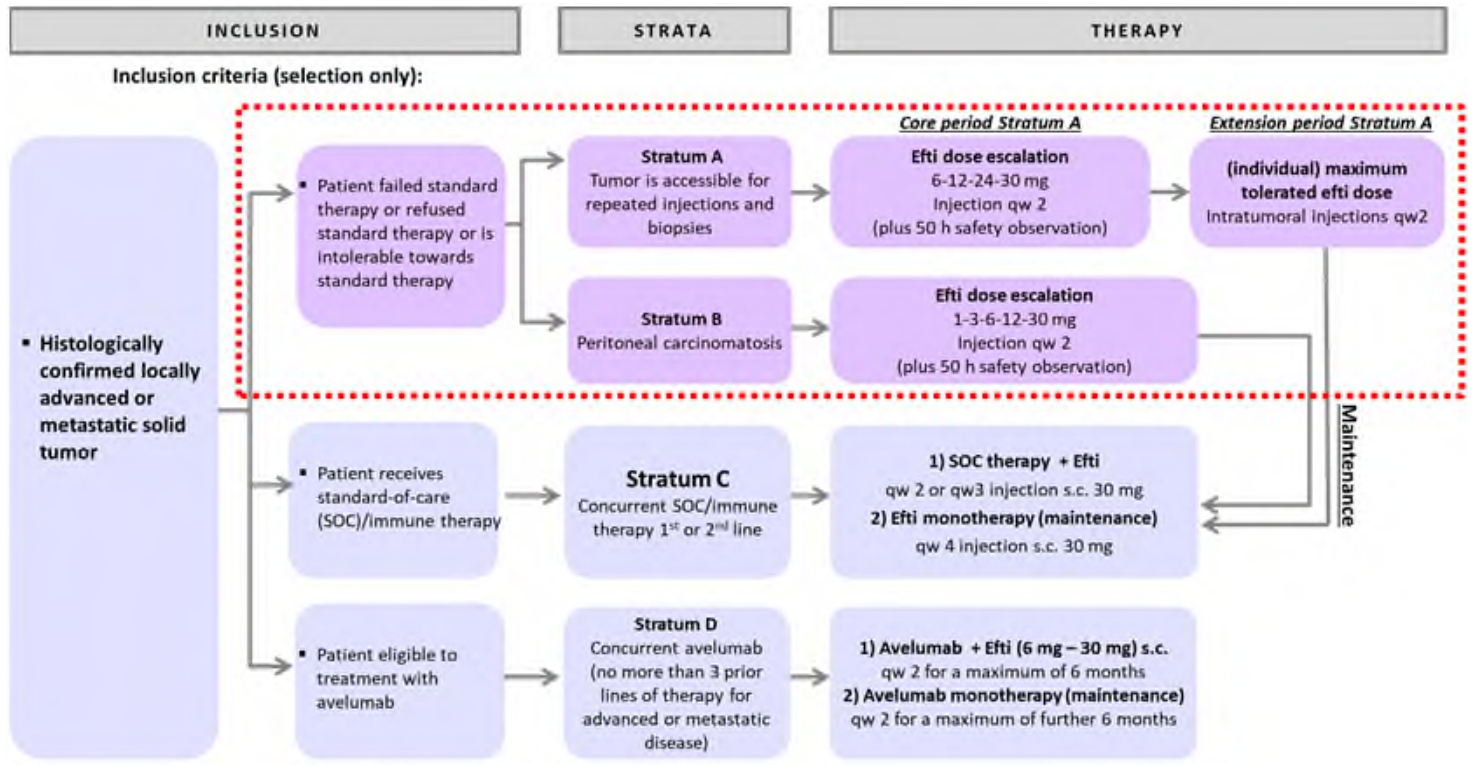

INCLUSION
STRATA
THERAPY
Inclusion criteria (selection only):
Core period Stratum A
Extension period Stratum A
Histologically confirmed locally advanced or metastatic solid tumor
Patient failed standard therapy or refused standard therapy or is intolerable towards standard therapy
Stratum A
Tumor is accessible for repeated injections and biopsies
Efti dose escalation
6-12-24-30 mg
Injection qw 2
(plus 50 h safety observation)
(individual) maximum tolerated efti dose
Intratumoral injections qw2
Stratum B
Peritoneal carcinomatosis
Efti dose escalation
1-3-6-12-30 mg
Injection qw 2
(plus 50 h safety observation)
Maintenance
Patient receives standard-of-care (SOC)/immune therapy
Stratum C
Concurrent SOC/immune therapy 1st or 2nd line
1) SOC therapy + Efti
qw 2 or qw3 injection s.c. 30 mg
2) Efti monotherapy (maintenance)
qw 4 injection s.c. 30 mg
Patient eligible to treatment with avelumab
Stratum D
Concurrent avelumab (no more than 3 prior lines of therapy for advanced or metastatic disease)
1) Avelumab + Efti (6 mg – 30 mg) s.c.
qw 2 for a maximum of 6 months
2) Avelumab monotherapy (maintenance)
qw 2 for a maximum of further 6 months

Methods

This is an investigator-initiated study with currently 4 strata (St): i.t. (A) or i.p. efti (B); s.c. efti with SOC (C) or combined with avelumab (D) (Figure 1). Here we focused on St A/B. In St A, patients (pts) received i.t. injections with efti escalation 6-12-24-30mg in cohort 1 and the maximum tolerated dose (MTD) in cohort 2. In St B, pts with peritoneal carcinomatosis received i.p. efti (dose escalation 1-3-6-12-30mg in cohort 1 and MTD in cohort 2). In both strata pts with a benefit after the last injection were offered s.c. efti for up to 52 weeks. Treatment was accompanied by translational analysis of selected immune markers. Main endpoint was safety.

Results

Recruitment has been completed with 8 pts treated in St A (cohort 1: 3 pts. [2 gastric cancer, 1 peritoneal mesothelioma]; cohort 2: 5 pts [cancer of head & neck, colon {2}, papilla, lung]) and 4 pts in St B cohort 1 (2 gastric, 2 colon cancer) (Table 1). Final analysis was performed. No dose limiting toxicities occurred. 14 serious adverse events (SAEs) have been reported: 8 in St A (2 in 1 pt of cohort 1, 6 in 4 pts of cohort 2) and 6 in 3 pts of St B cohort 1 (Table 2, Table 3). 1 SAE in cohort 2 of St A was related to study procedure (sudden death NOS grade 5). 1 AESI (chills grade 3) probably related to efti occurred in St A cohort 1. Of the heavily pretreated pts, 5 had stable disease (SD) (3 gastric, 2 colon cancer), 5 progressive disease (RECIST) and 2 clinical progression. 2 of the SD pts. had PFS of 3mo (1 gastric St A; 1 colon St B) and 2 SD pts. had PFS of 4mo (2 gastric St B) (Table 1). The SD gastric cancer pt of St A with PFS of 3 mo had an OS of 28 mo with increase of PD-L1 in the immune cells from 20% at baseline (BL) to 30% (D29 + D71) (Figure 2, Figure 3). 3 SD pts with gastric cancer showed high CD45, CD163 expression at BL (tissue). Blood cytokine profile: 1 SD St B pt (001-002) showed a significant increase in IFN ɣ, 4h - 24h after inj. measured on D1/D29, combined with a moderate increase in CD4 and CD8 cells (Figure 4). Increase in IFN ɣ with additional NK cells was also seen in 1 SD St A pt (001-001).

Table 1: Patient overview

Pat-ID	Stratum / Cohort	Indication	Last prior therapy	PD-L1 staining / MSI / Molecular markers	No of efti injections	Injected lesion	Max dose [mg]	Best response	PFS (months)	OS (months)
001-001	A / 1	Gastric cancer	FOLFIRI / 2nd line	TPS 10%; CPS 15%	7	Stomach	30	SD	3	28
001-004	A / 1	Peritoneal mesothelioma	Cis-platin / pemetrexed / 3rd line	No data available	4	Abdominal wall	30	PD	1	3
001-005	A / 1	Gastric cancer	Irinotecan + ramucirumab / 3rd line	MSS	4	Abdominal wall	30	PD	1	2
001-007	A / 2	Head and neck cancer	Docetaxel / 3rd line	No data available	4	Liver	30	PD	2	14
001-008	A / 2	Colon cancer	FOLFOX / 6th line	MSS / Ras mt	4	Liver	30	PD	2	2
001-010	A / 2	Papilla carcinoma	FLO / 4th line	PD-L1 neg / MSS / Her2+	4	Liver	30	PD	1	1
001-012	A / 2	Colon cancer	FOLFIRI / 6th line	MSS / RAS wt - BRAF wt	1	Liver	30	missing (clinical progression)	2	2
001-015	A / 2	Lung cancer	Gemcitabine mono / 5th line	TPS 50%	1	Lung	30	missing (clinical progression)	1	1
001-002	B / 1	Colon cancer	Capecitabine / 2nd line	No data available	6	Peritoneum	30	SD	3	20
001-006	B / 1	Gastric cancer	FOLFIRI / 3rd line	No data available	6	Peritoneum	30	SD	4	4
001-013	B / 1	Colon cancer	FOLFOX + bevazizumab / 3rd line	MSS / RAS wt / BRAF wt	1	Peritoneum	1	SD	0	0
001-016	B / 1	Gastric cancer	Ramucirumab / 2nd line	No data available	5	Peritoneum	30	SD	4	4

Table 2: Summarized SAEs by patients

SAE	Stratum A Cohort 1 Escalation cohort n=3 (%)	Stratum A Cohort 2 Consolidation cohort n=5 (%)	Stratum A Total n=8 (%)	Stratum B Cohort 1 Escalation cohort n=4 (%)
Patients with at least one SAE	1 (33%)	4 (80%)	5 (63%)	3 (75%)
Patients with at least one SAE with relation to study treatment	0 (0%)	1 (20%)	1 (13%)	0 (0%)
Patients with at least one AESI (related to study treatment)	1 (33%)	0 (0%)	1 (13%)	0 (0%)

Table 3: Serious adverse events (irrespective of relationship to study drug)

	Stratum A Cohort 1 Escalation cohort n=3 (%)		Stratum A Cohort 2 Consolidation cohort n=5 (%)		Stratum A Total n=8 (%)		Stratum B Cohort 1 Escalation cohort n=4 (%)	
Serious adverse event	**G3**	**G5**	**G3**	**G5**	**G3**	**G5**	**G3**	**G5**
Colonic hemorrhage	1 (33%)				1 (13%)			
Gastric hemorrhage		1 (33%)				1 (13%)		
Bile duct stenosis			1 (20%)		1 (13%)			
Ascites			1 (20%)		1 (13%)			
Fatigue			1 (20%)		1 (13%)			
Pleural infection				1 (20%)		1 (13%)		
Sudden death NOS				1 (20%)		1 (13%)		
Aspiration				1 (20%)		1 (13%)		
Ileus							2 (50%)	
Thromboembolic event							1 (25%)	
Tumourpain due to tumour progression								1 (25%)
Multi-organ failure								1 (25%)
Vomiting							1 (25%)	

Table 4: Most common adverse events (irrespective of relationship to study drug)

	Stratum A Total n=8 (%)				Stratum B Cohort 1 (Escalation cohort) n=4 (%)			
Most common AEs	**G1/G2**	**G3**	**G4**	**G5**	**G1/G2**	**G3**	**G4**	**G5**
Alkaline phosphatase increased	2 (25.0%)							
Anemia		2 (25.0%)						
Ascites		1 (12.5%)			1 (25.0%)			
Chills	1 (12.5%)	1 (12.5%)			1 (25.0%)			
GGT increased		1 (12.5%)	1 (12.5%)					
Hypertension		2 (25.0%)						
Hypoalbuminemia	2 (25.0%)							
Ileus						2 (50.0%)		
Malaise	1 (12.5%)				2 (50.0%)			
Nausea		1 (12.5%)			1 (25.0%)	1 (25.0%)		
Pain	3 (37.5%)	1 (12.5%)			1 (25.0%)	1 (25.0%)		
Vomiting		1 (12.5%)			1 (25.0%)	1 (25.0%)		

Table 5: Treatment related AEs

	Stratum A Total n=8 (%)				Stratum B Cohort 1 (Escalation cohort) n=4 (%)			
Adverse reaction	**G1/G2**	**G3**	**G4**	**G5**	**G1/G2**	**G3**	**G4**	**G5**
Abdominal pain					1 (25.0%)			
Chills	1 (12.5%)	1 (12.5%)			1 (25.0%)			
Constipation					1 (25.0%)			
Diarrhea					1 (25.0%)			
Dizziness	1 (12.5%)							
Fever	1 (12.5%)				1 (25.0%)			
Hypoxia	1 (12.5%)							
Nausea		1 (12.5%)						
Pain	1 (12.5%)							
Pain of skin	1 (12.5%)							
Pruritus	1 (12.5%)							
Sudden death NOS				1 (12.5%)				
Vomiting		1 (12.5%)						

Translational Data

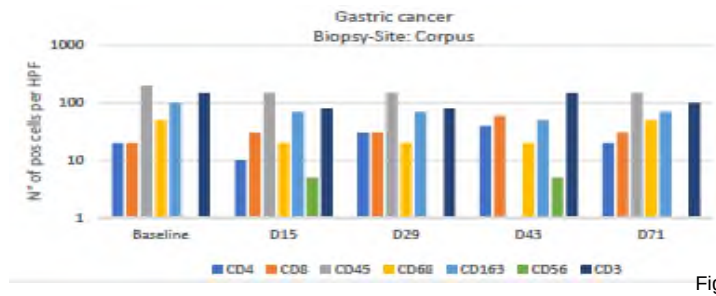

Gastric cancer
Biopsy-Site: Corpus
N° of pos cells per HPF
Baseline
D15
D29
D43
D71
CD4
CD8
CD45
CD68
CD163
CD56
CD3

Figure 2 (pt 001-001)

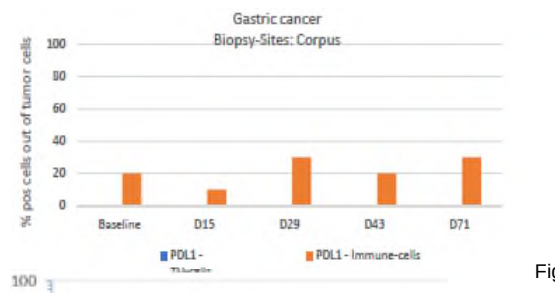

Gastric cancer
Biopsy-Sites: Corpus
% pos cells out of tumor cells
Baseline
D15
D29
D43
D71
PDL1 - Immune-cells

Figure 3 (pt 001-001)

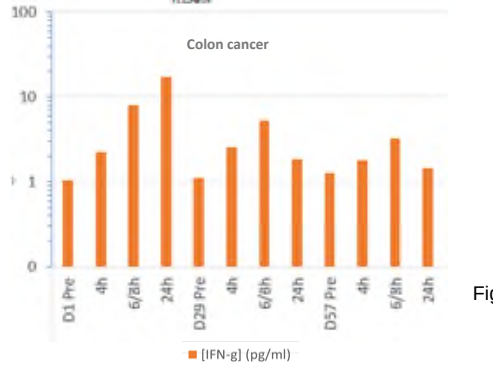

Colon cancer
D1 Pre
4h
6/8h
24h
D29 Pre
4h
6/8h
24h
D57 Pre
4h
6/8h
24h
[IFN-g] (pg/ml)

Figure 4 (pt 001-002)

Conclusion

Intratumoral and intraperitoneal administration of efti up to 30 mg is feasible, although technically challenging, and displayed signals of clinical and cytokine activity.

First author contact information:
Salah-Eddin Al-Batran, albatran@ikf-khnw.de

First author conflicts of interest:
Resarch grant IMMUTEP

Study management contact information:
Regina Eickhoff, eickhoff.regina@ikf-khnw.de

Study sponsored (grant/IMP) by:
Immutep GmbH

Study identifiers:
EudraCT-No.: 2016-002309-20
Clinicaltrials.gov: NCT03252938
ID 1033P ESMO 2020